Mandibular Bone Height Changes Associated with Screw-Retained Implant-Supported Prostheses with Distal Cantilevers: A Long Term Follow Up Analysis

Thomas J. Balshi, DDS, PhD, FACP*; Glenn J. Wolfinger, DMD, FACP*; Matilda Dhima, DMD**; Vicki C. Petropoulos, DMD, MS**; Stephen F. Balshi, MBE*

* PI Dental Center at the Institute for Facial Esthetics, Fort Washington, PA

** University of Pennsylvania, Philadelphia, PA

Introduction: Residual ridge resorption occurs following extractions or loss of teeth. The rate of resorption is variable and considered a multifaceted process including metabolic, anatomic and biomechanical factors. When patients are restored with a removable denture, the residual ridge resorption process does not stop. However, it has been reported in some studies that patients rehabilitated with an implant-supported fixed cantilever prosthesis can not only stop the process, but reverse it.[1,2] The longest follow up study available in the literature evaluated these bone changes up to 4 years.

The main purpose of this study was to evaluate the long term changes in bone height of mandibular screw-retained implant-supported prostheses with distal cantilevers and to determine if the reversal of residual ridge resorption in the posterior mandible is only short-term or if it is continued into the long-term management of the patient.

Methods: Panoramic radiographs for each patient were obtained at time of surgery and for two sequential exam visits with varying follow up times. Digital panoramic radiographs were magnified using a computer system with calibrated and known magnification error which was used to convert the pixels into millimeters. The actual implant length and bone height at time of surgery were used as baselines to correct for variation in implant radiographic magnification. Changes in bone height were evaluated between each follow up visit as well as from time of surgery to last follow up. Statistical tests, means, standard deviations, a two way analysis of variation (ANOVA) for the measurements obtained at each exam, and piecewise linear regression model were performed to evaluate the mean changes in bone height between each exam. Characteristics of trends in bone height changes for patient gender were also evaluated.

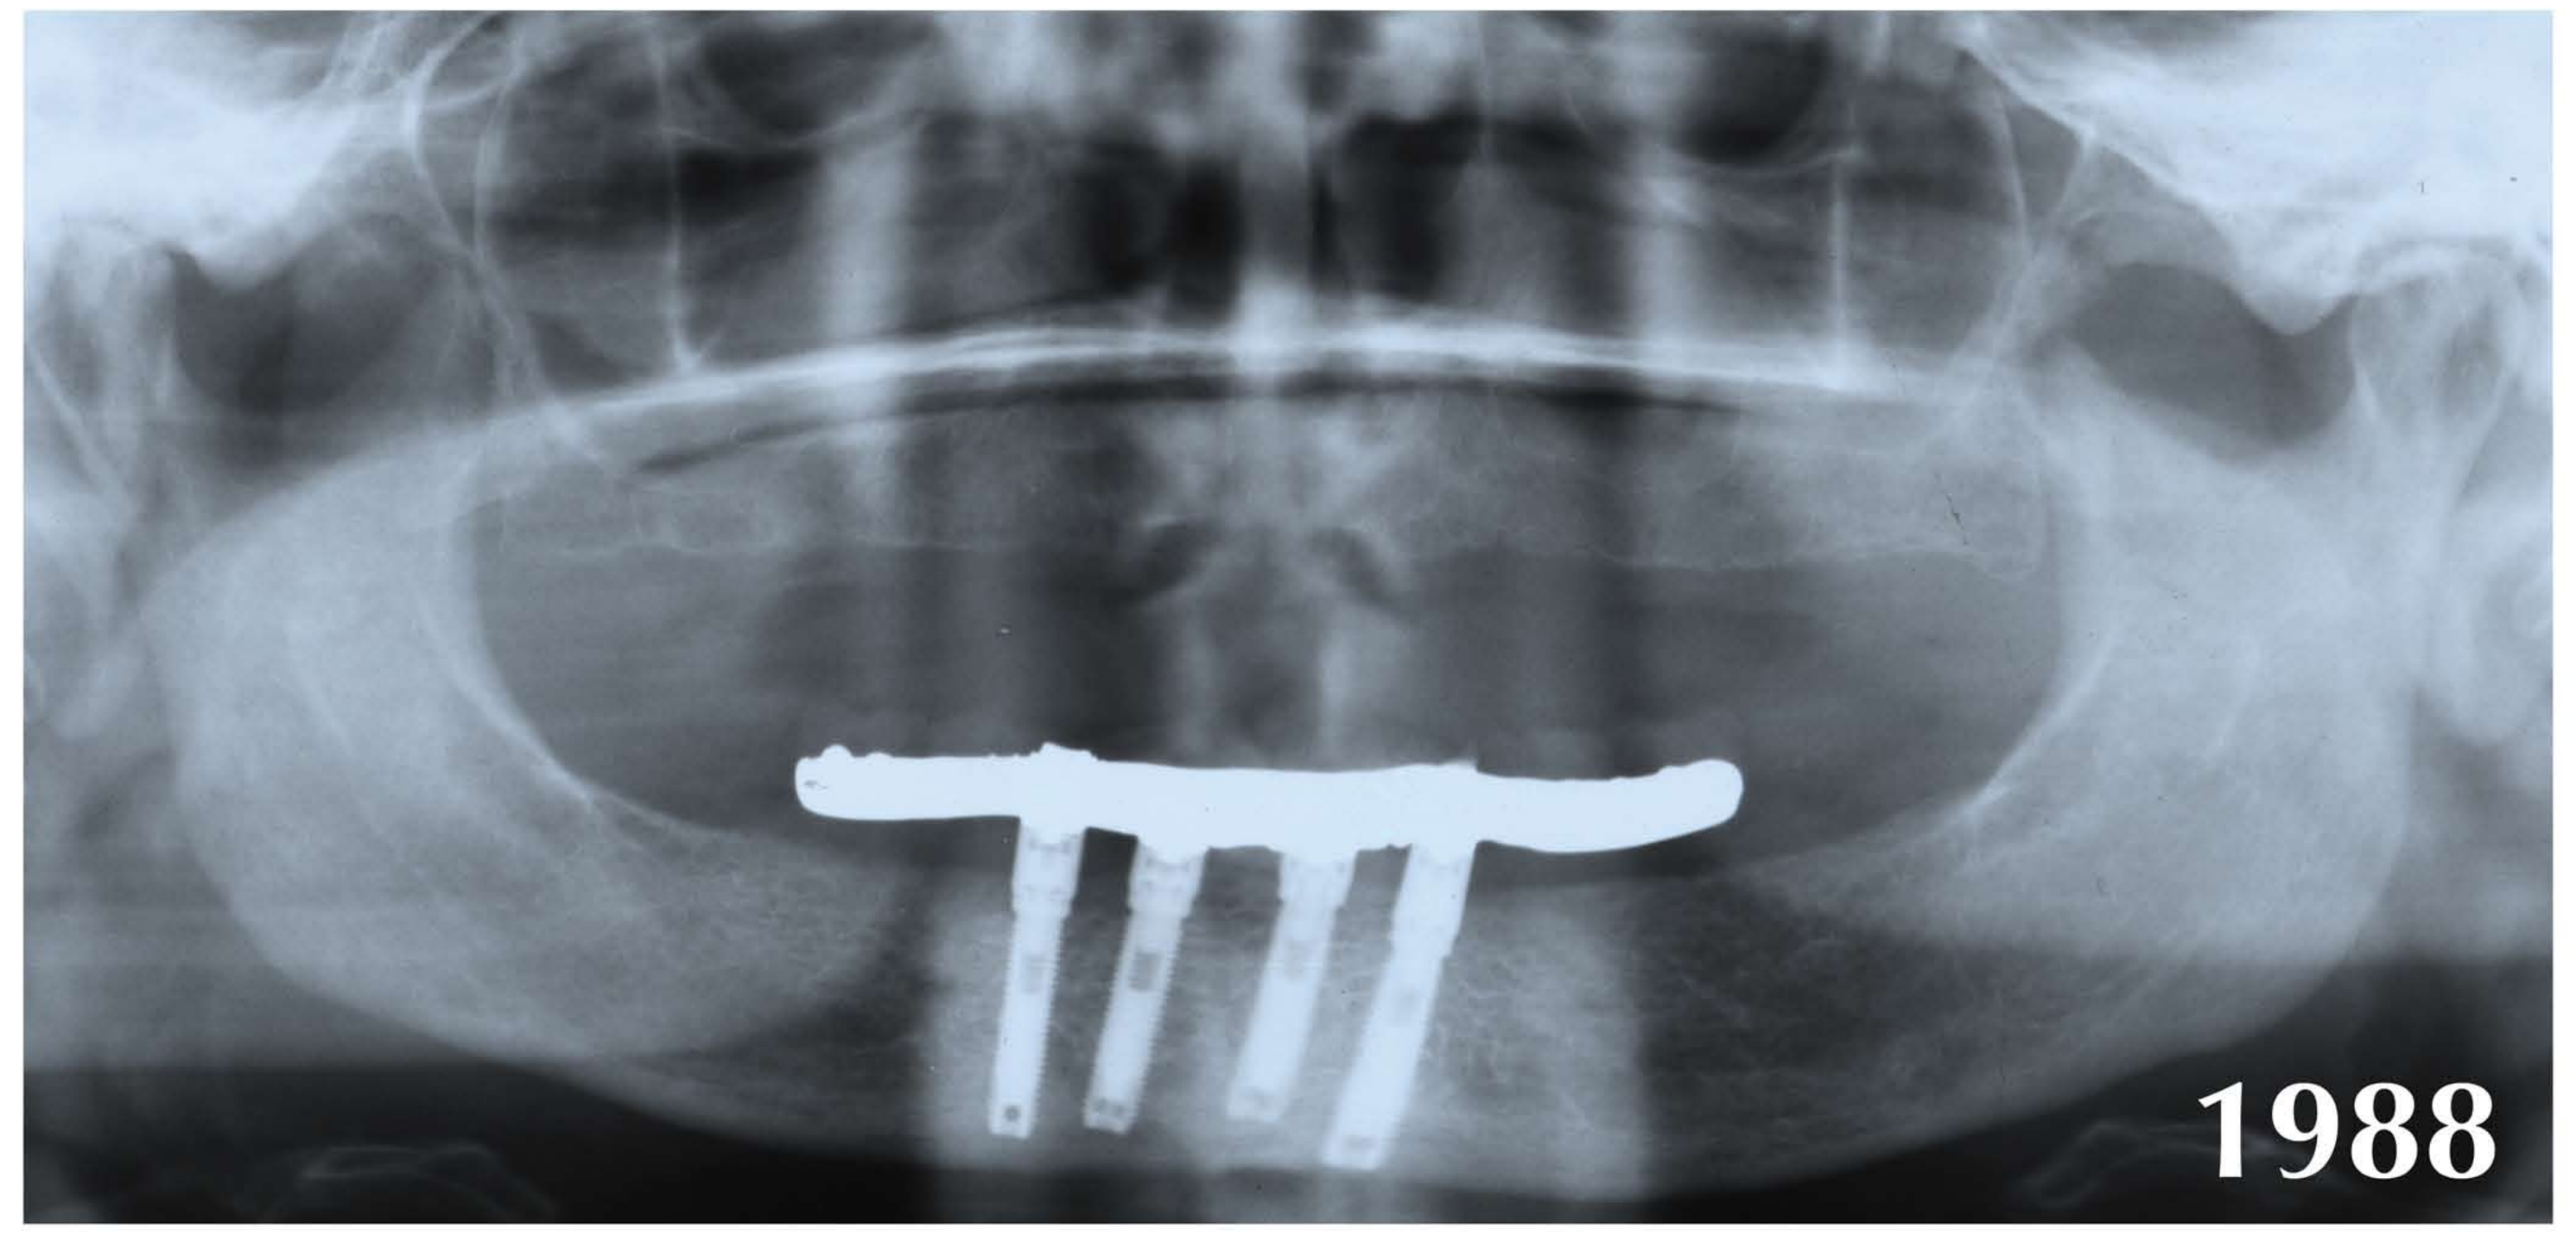
1988

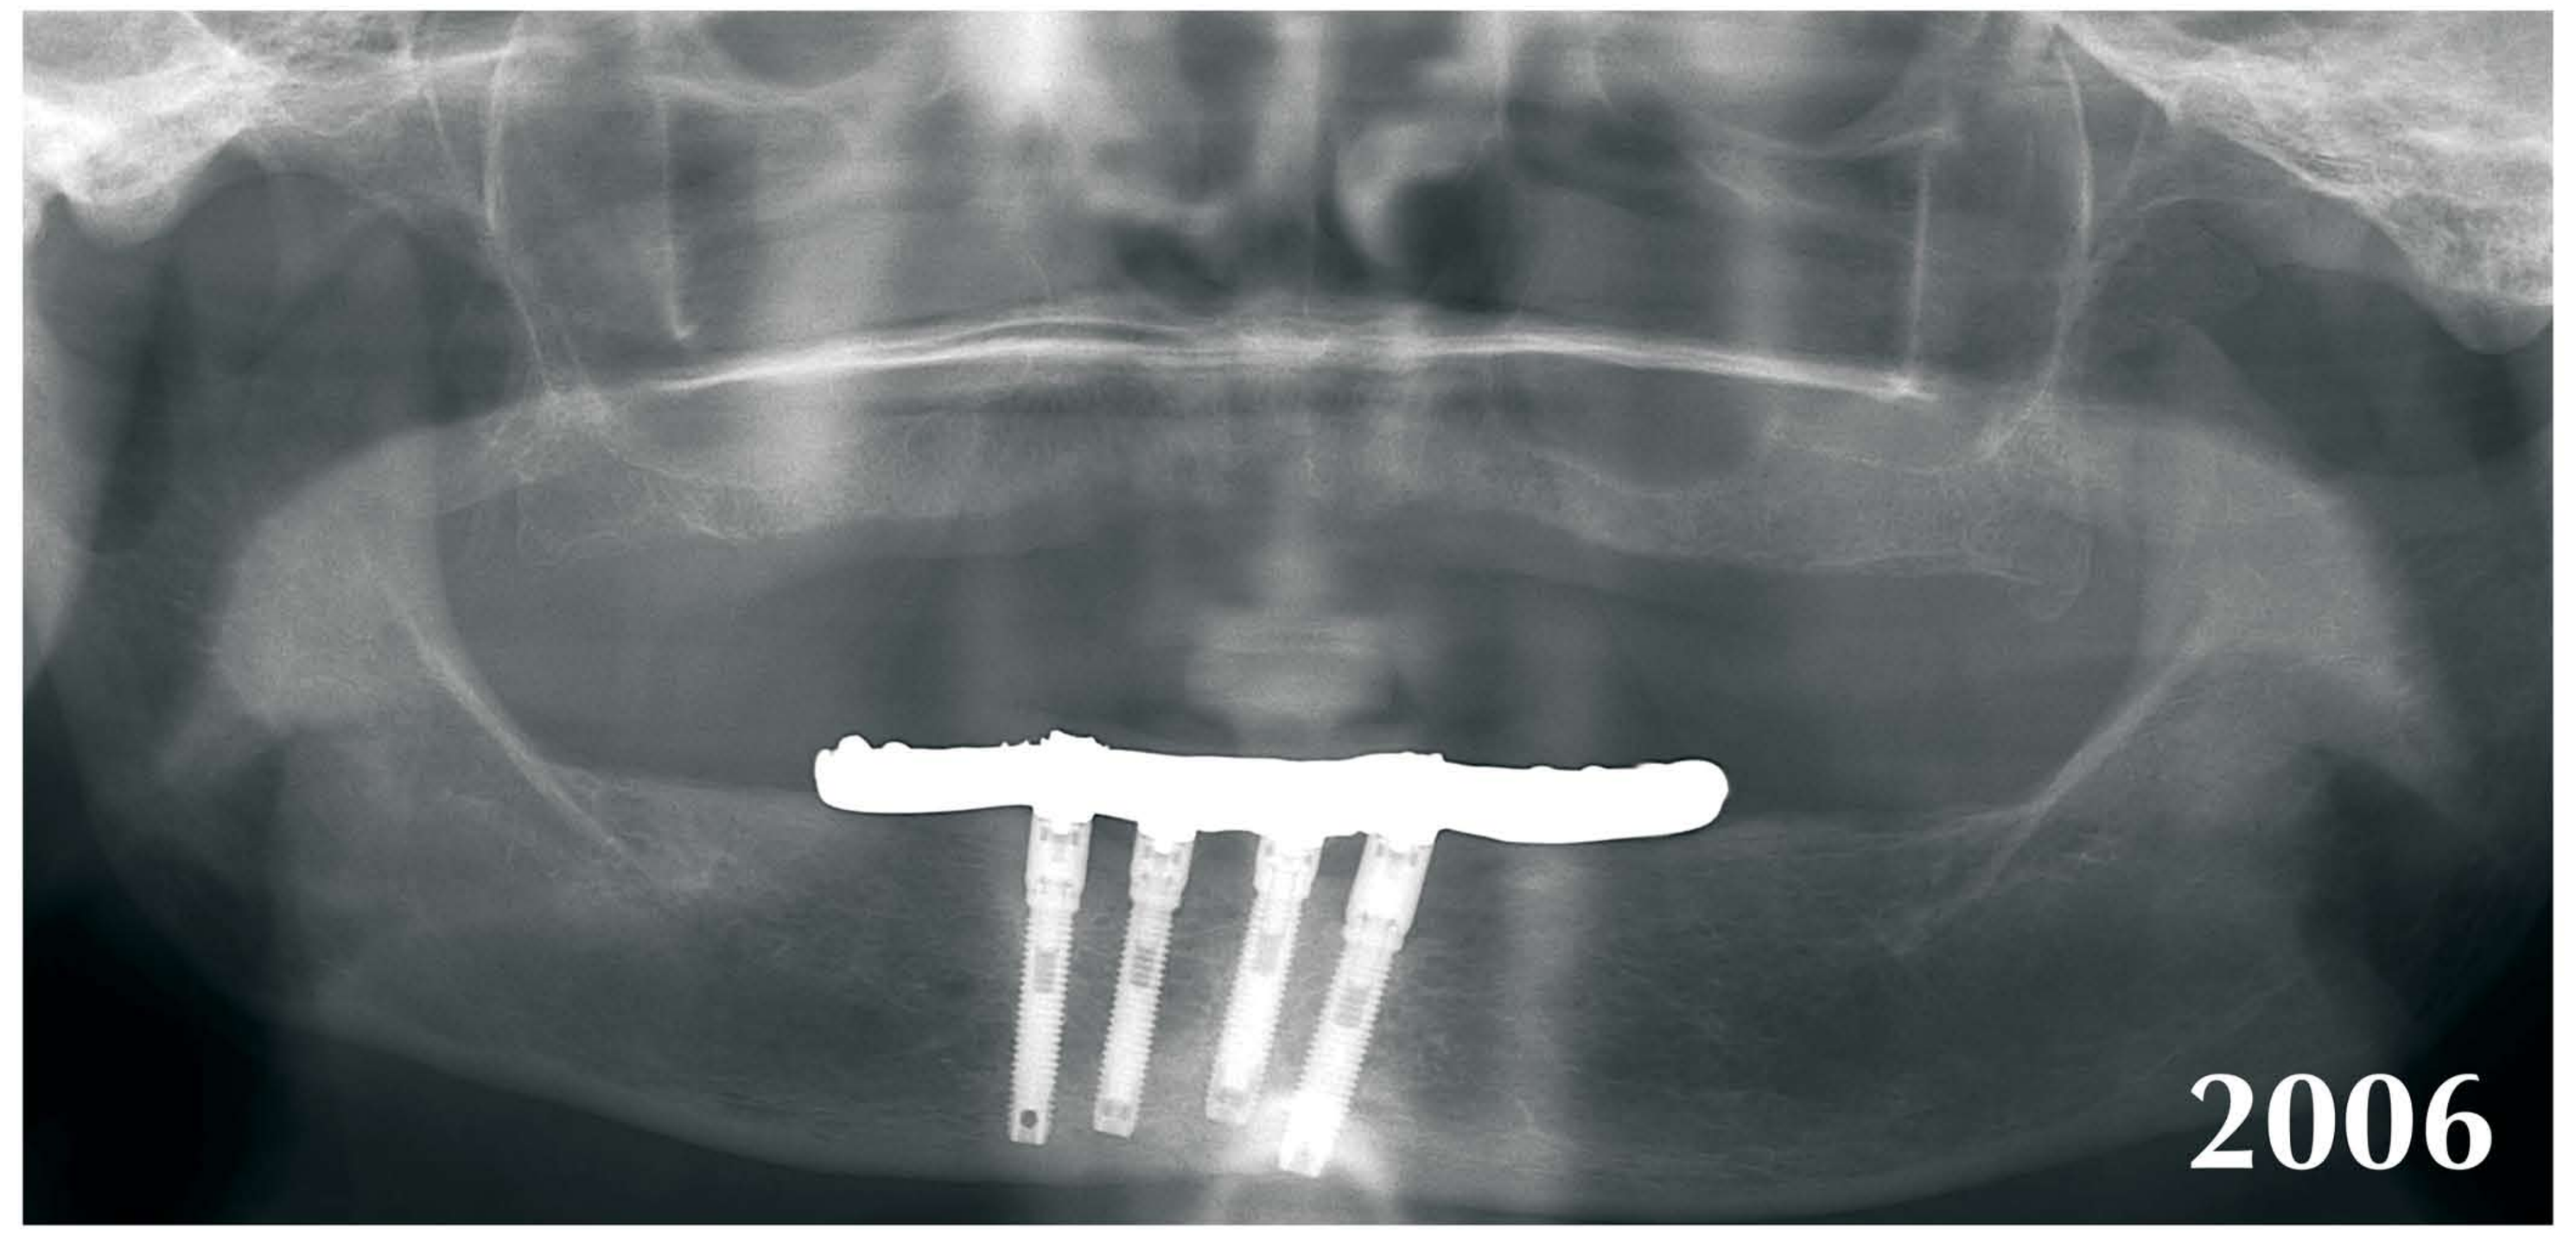
2006

Figure 1: Total Study Population

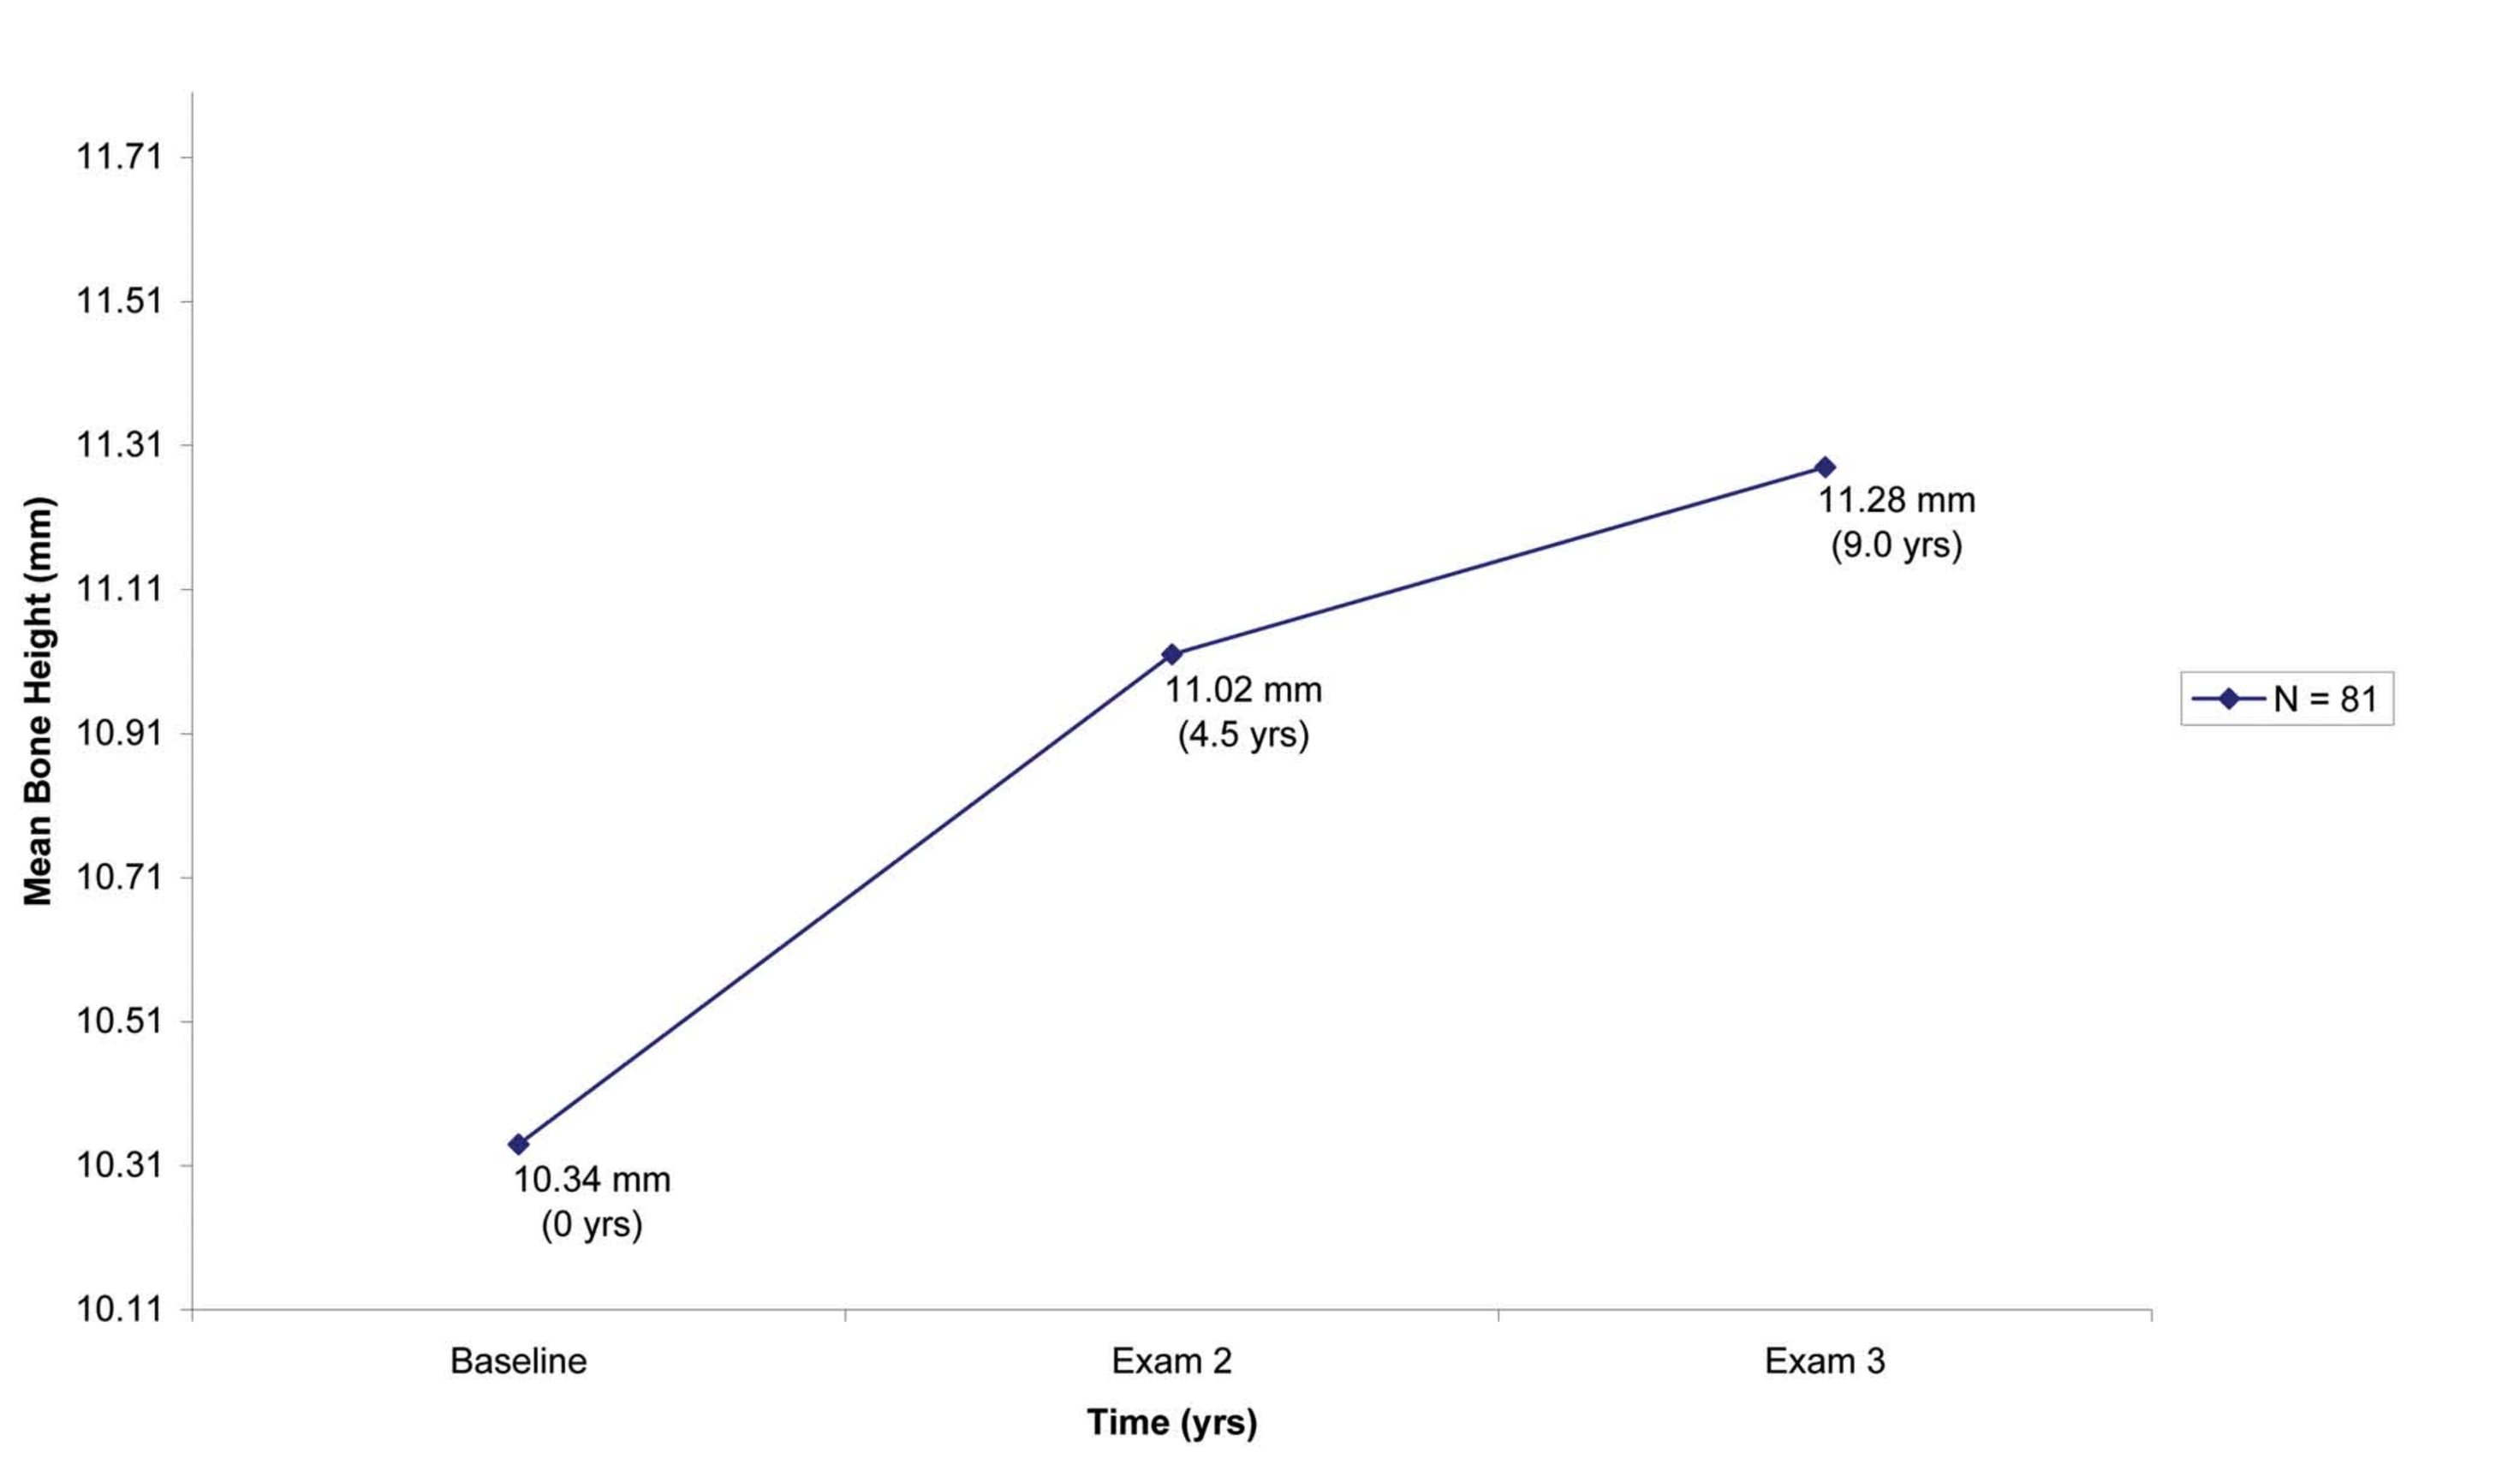
11.71
11.51
11.31
11.11
10.91
10.71
10.51
10.31
10.11
Mean Bone Height (mm)
10.34 mm (0 yrs)
11.02 mm (4.5 yrs)
11.28 mm (9.0 yrs)
N = 81
Baseline
Exam 2
Exam 3
Time (yrs)

LONGEST FOLLOW-UP TIME, MALE = 15 YEARS	
YEAR	BONE HEIGHT
1989	7.14 mm
2002	7.32 mm
2004	7.50 mm

* patient expericed an overall bone height gain of 0.45 mm over a period of 15 years

Acknowledgments: The authors sincerely appreciate the contributions from Marjorie K. Jeffcoat, DMD for advice on data acquisition and assistance with acquiring such data; Christine Raines for assistance with image preparation; and John William Fitzpatrick for assistance with design and layout of this poster.

.

Figure 2: Gender Comparison

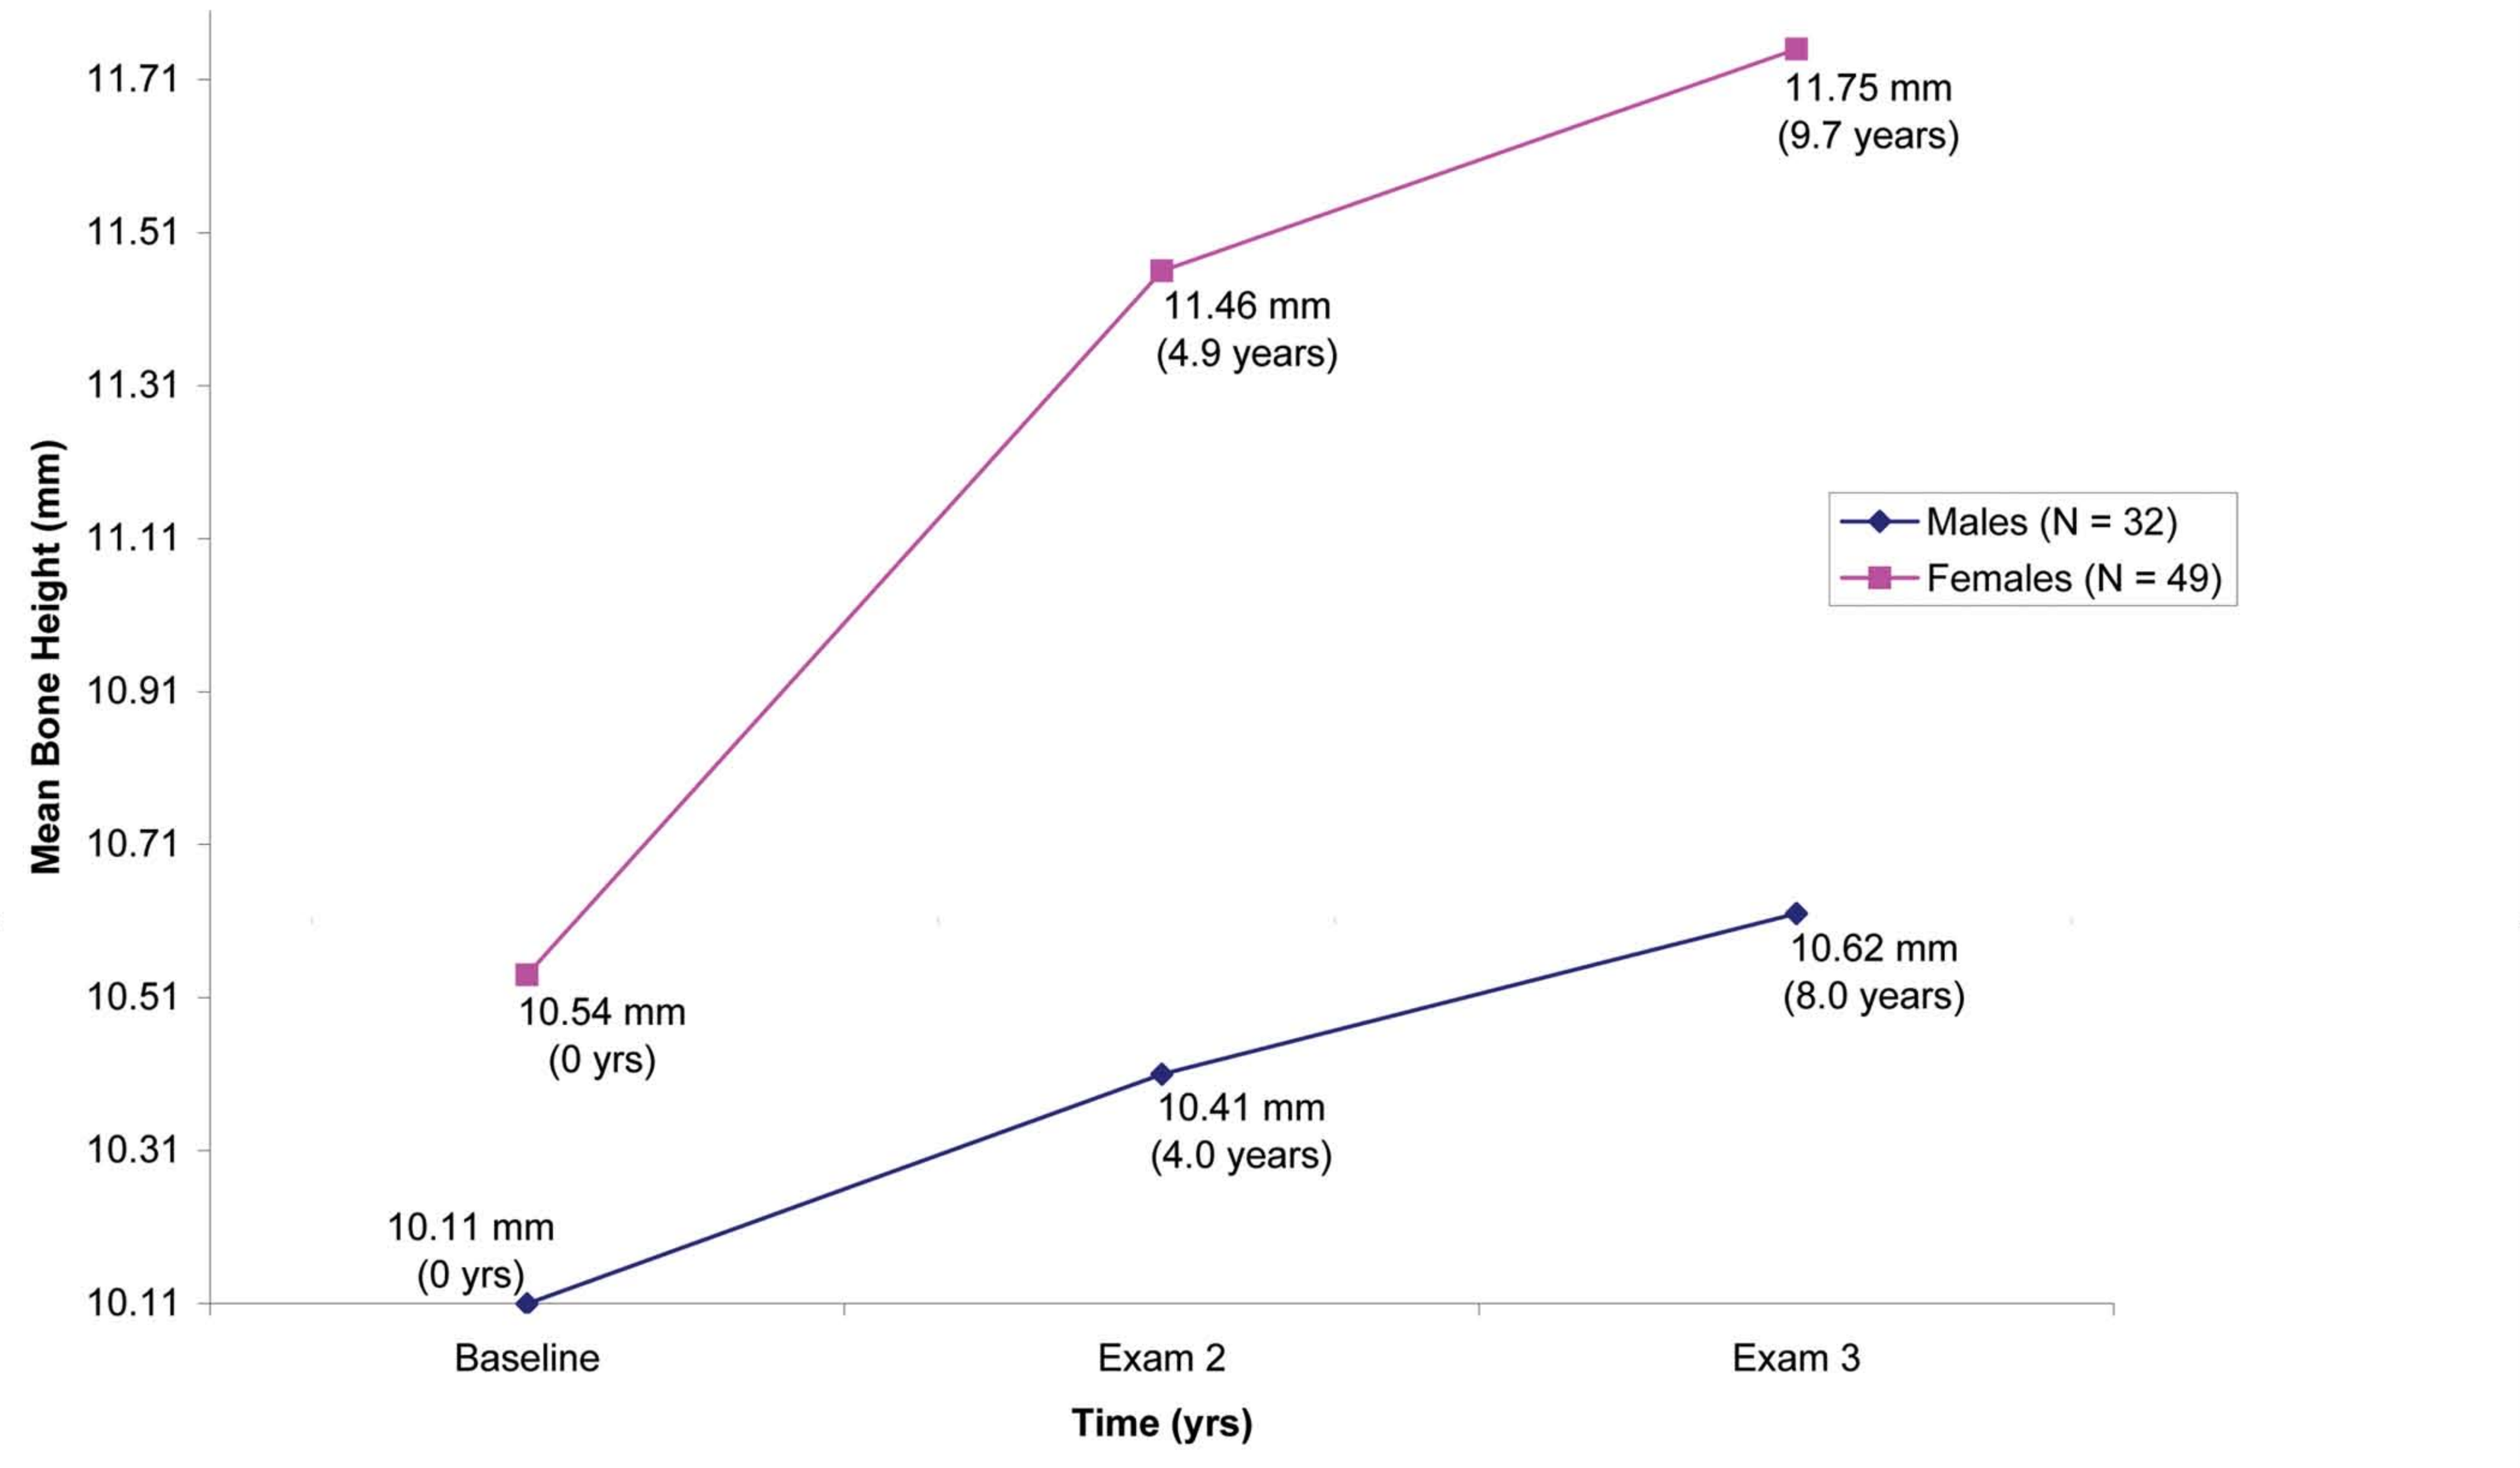
11.71
11.51
11.31
11.11
10.91
10.71
10.51
10.31
10.11
Mean Bone Height (mm)
10.54 mm (0 yrs)
11.46 mm (4.9 years)
11.75 mm (9.7 years)
10.11 mm (0 yrs)
10.41 mm (4.0 years)
10.62 mm (8.0 years)
Males (N = 32)
Females (N = 49)
Baseline
Exam 2
Exam 3
Time (yrs)

LONGEST FOLLOW-UP TIME, FEMALE = 19 YEARS	
YEAR	BONE HEIGHT
1988	9.42 mm
1993	11.38 mm
2007	10.71 mm

* patient experienced both bone gain and bone loss over a period of 19 years with an overall gain of 1.29 mm

References:

1) Wright PS, Glantz P-O, Randow K, Watson RM. The effects of fixed and removable implant-stabilised prostheses on posterior mandibular residual ridge reorption. Clin Oral Implants Res 2002;13:169-74.

2) Reddy MS, Geurs NC, Wang I-C, Liu P-R, Hsu Y-T, Jeffcoat RL, Jeffcoat MK. Mandibular Growth Following Implant Restoration: Does Wolff's Law Apply to Residual Ridge Resorption? Int J Periodontics Restorative Dent 2002;22:315-21.

Results: This study evaluated changes in mandibular bone height in 81 patients treated with mandibular screw-retained implant-supported prostheses with a distal cantilever. These prostheses were supported by four, five or six implants. As illustrated in **Figure 1**, mean average bone height distal to the last implant at time of implant placement was 10.34 mm. Average time between implant placement and first follow up exam was 4.5 years. Mean bone height changes from time of implant placement to first follow up exam show a mean bone height gain of 0.68 mm. Average time between the first and second follow up exams was also 4.5 years resulting in an average of 9.0 years follow up. Mean bone height changes from the first follow up exam to the second follow up exam was an additional gain of 0.26 mm.

Figure 2 illustrates the comparison of the 81 patients divided into gender. The mean baseline measurement for females was 0.43 mm greater than males. A statistically significant amount of bone height gain was noticed in the female population between the first and second exam when compared to the males. This statistically significant difference was not seen from the second exam to the third exam between males and females. All but 2 females experienced bone gain between baseline and the second exam. The greatest bone height gain noticed was 3.61 mm in a female examined 2 years after baseline.

Conclusion: This study shows that although overall bone growth is associated with mandibular screw-retained implant-supported prostheses with distal cantilevers, both bone loss and bone growth may occur in the same patient throughout time. Further evaluation is required to see if bone height change is a function of the amount of baseline bone height.